A Safe and Highly Potent αPD1-IL2 Fusion (AWT020) that Decouples the Efficacy and Toxicity of IL-2 Therapy

Anwita Biosciences
Fan Ye fye@anwitabio.com
Ziyang Zhong zzhong@anwitabio.com
300 Industrial Rd. San Carlos, CA 94070

Fan Ye, Jianing Huang, Lili Cheng, Zoey Huang, Sandra Chen, Danny Huang, Michael Hua, Ella Li, Jenny Jiang, Hanna Lin, Xin Wang, Bella Yue, Shirley Shi, Henry He, Karen Yan, Jiamin Wang, Binfeng Lu, Ziyang Zhong

SITC 2022 November 8-12 Boston Massachusetts
Abstract Number.: 1104

INTRODUCTION

Interleukin 2 (IL-2) is one of the most important cytokines for cancer immunotherapy[1]. The clinical efficacy of IL-2 had been proven in the treatment of metastatic renal cell carcinoma and metastatic melanoma with overall response rate of 15-25% and the response is generally dose dependent[2,3]. However, high dose IL-2 treatment is associated with severe adverse effects such as vascular leak syndrome (VLS), which affects multiple organs and greatly limits the therapeutic potential of IL-2[4].

The toxicity of IL-2, over many years of scientific research, has been attributed to its binding to CD25, or IL-2Rα[5,6]. Recently, a number of commonly named No-α-IL2 variants with abolished IL-2Rα binding, were demonstrated to have an improved safety profile in the early phase of clinical studies. Compared to WT IL-2, No-α-IL2 has less stimulation to eosinophils and regulatory T cells (Treg), while it still universally activates all the IL-2Rβγ expressing immune cells, including T cells and NK cells[7].

To further enhance the specificity of IL-2, we engineered a fusion protein comprised of an anti-PD-1 antibody and a potency optimized No-α-IL2 (AWT020). AWT020 selectively stimulated the expansion of tumor infiltrating T cells while avoiding NK expansion, demonstrating a superb safety profile and excellent antitumor efficacies. It was also shown to be superior to an industrial leading αPD1-IL-2x as tested in multiple tumor models.

TARGET TUMORAL T CELLS BY DESIGN

PD1 Expression is Highly Specific to TIL | **PD1 Blockade Synergizes with IL-2** | **PD1 Dependent TIL Activation**

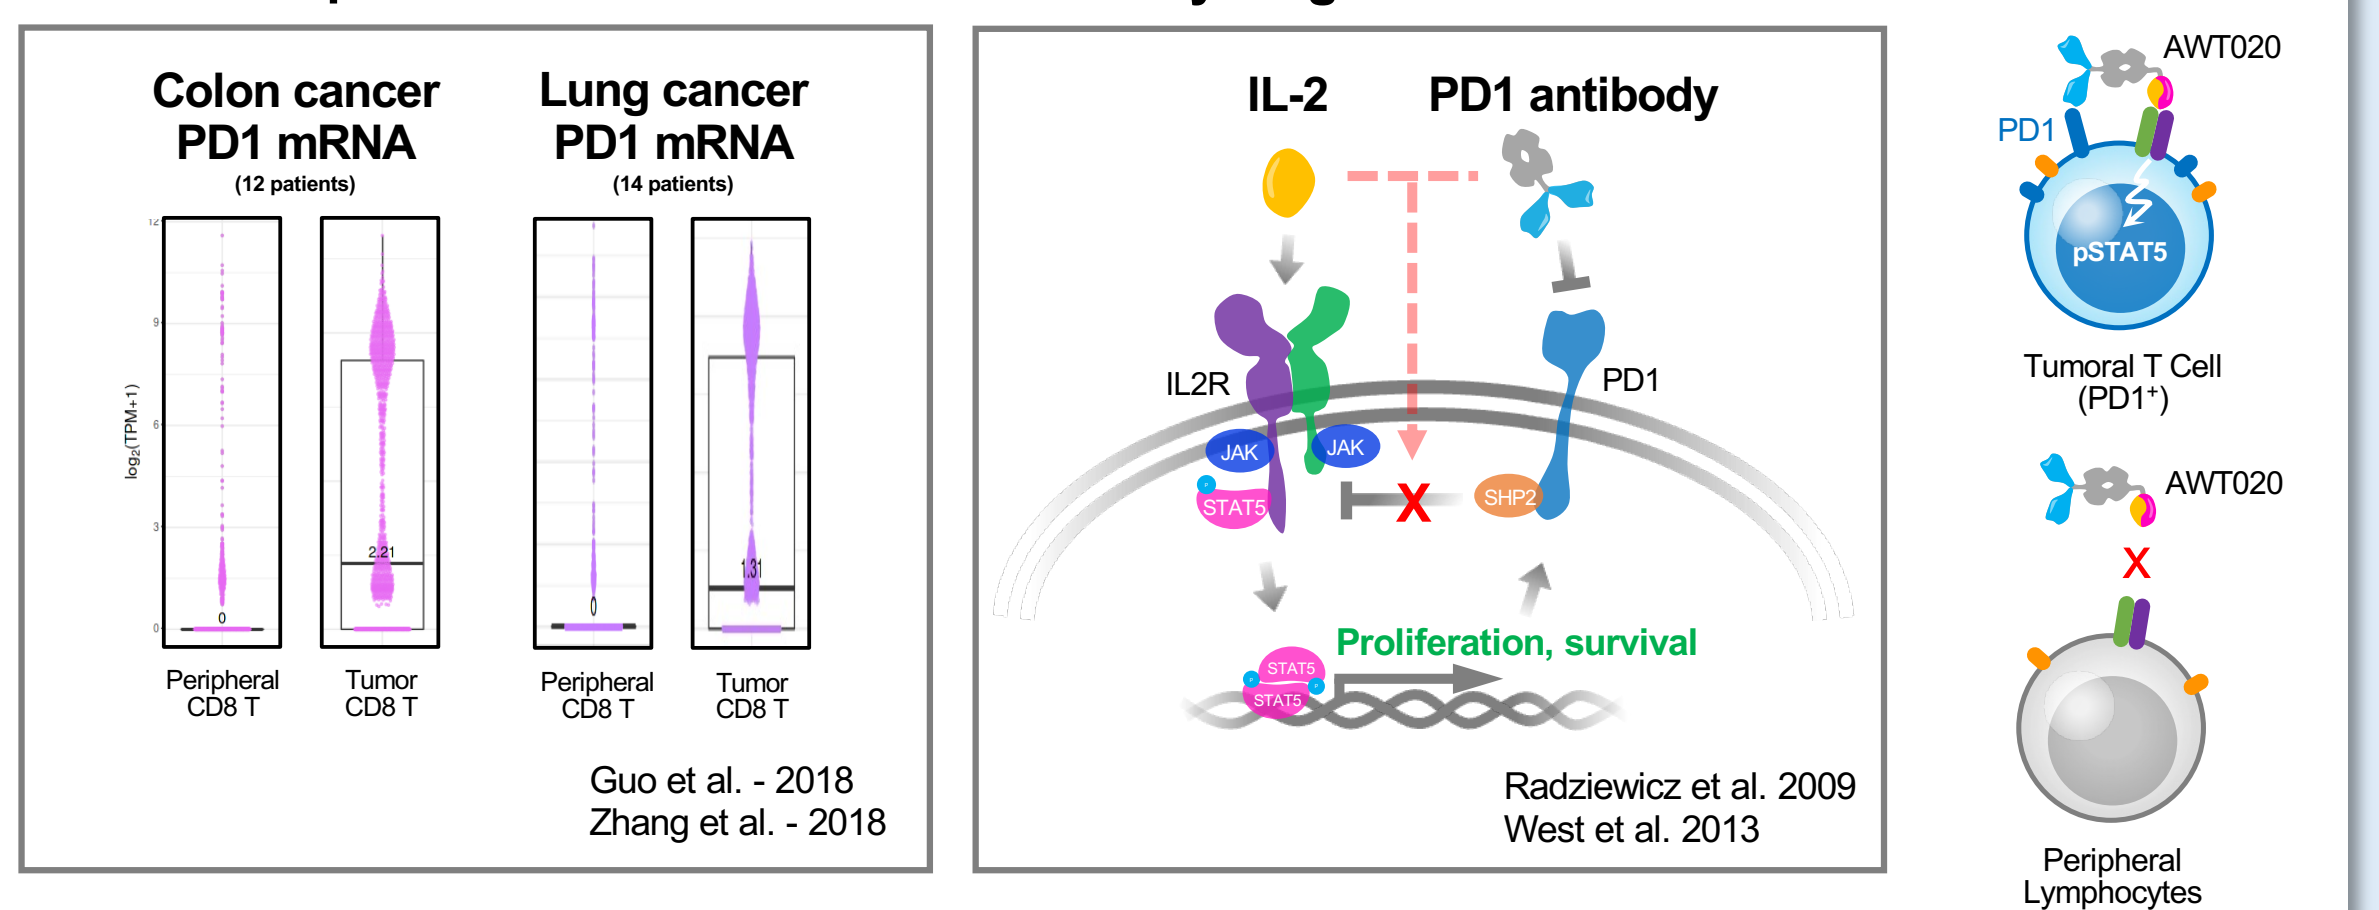
Colon cancer PD1 mRNA
(12 patients)
Lung cancer PD1 mRNA
(14 patients)
Peripheral CD8 T
Tumor CD8 T
Guo et al. - 2018
Zhang et al. - 2018
IL-2
PD1 antibody
IL2R
PD1
JAK
STAT5
SHP2
Proliferation, survival
Radziewicz et al. 2009
West et al. 2013
AWT020
PD1
pSTAT5
Tumoral T Cell (PD1+)
Peripheral Lymphocytes (PD1-)

Toxicity ↓ **Efficacy ↑**

AWT020 Selectively Activates PD-1+ T Cells

Potent and Selective Activator of STAT5 signaling

Potent and Selective Expander of PD1[high] Human CD3+ T Cells

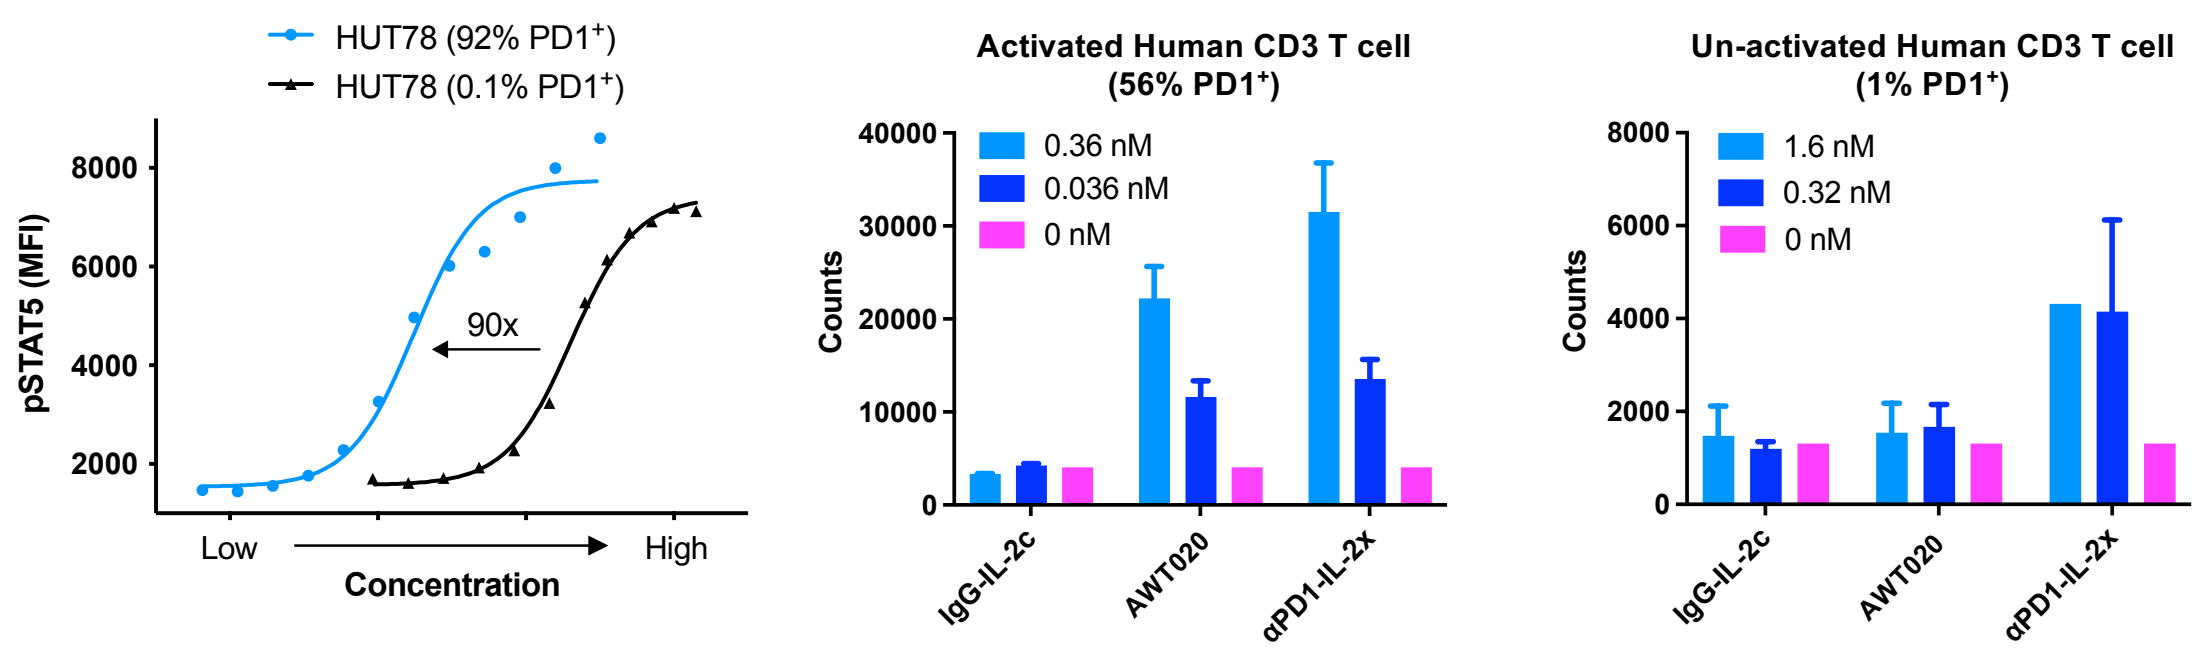
HUT78 (92% PD1+)
HUT78 (0.1% PD1+)
pSTAT5 (MFI)
90x
Low
High
Concentration
Activated Human CD3 T cell (56% PD1+)
0.36 nM
0.036 nM
0 nM
Counts
Un-activated Human CD3 T cell (1% PD1+)
1.6 nM
0.32 nM
0 nM
IgG-IL-2c
AWT020
αPD1-IL-2x

- AWT020 potently induced pSTAT5 signaling in PD1+ T cell lymphoma cells (HUT78)
- αPD1-IL-2x robustly enhanced the proliferation of both PD1[high] and PD1[low] human CD3+ T cells.
- AWT020 only induced the proliferation of PD1[high] human CD3+ T cells.
- Non-targeting IgG-IL-2c control showed no obvious induction of T cell proliferation.

SUPERIOR T CELL ANTITUMOR IMMUNITY

mAWT020 Is A Superior Antitumor Agent

Superior Antitumor Activity Than Single Agents Combined | Robust Immunological Memory From Complete Responders | Potent Antitumor Activity with Only 1 Dose of mAWT020

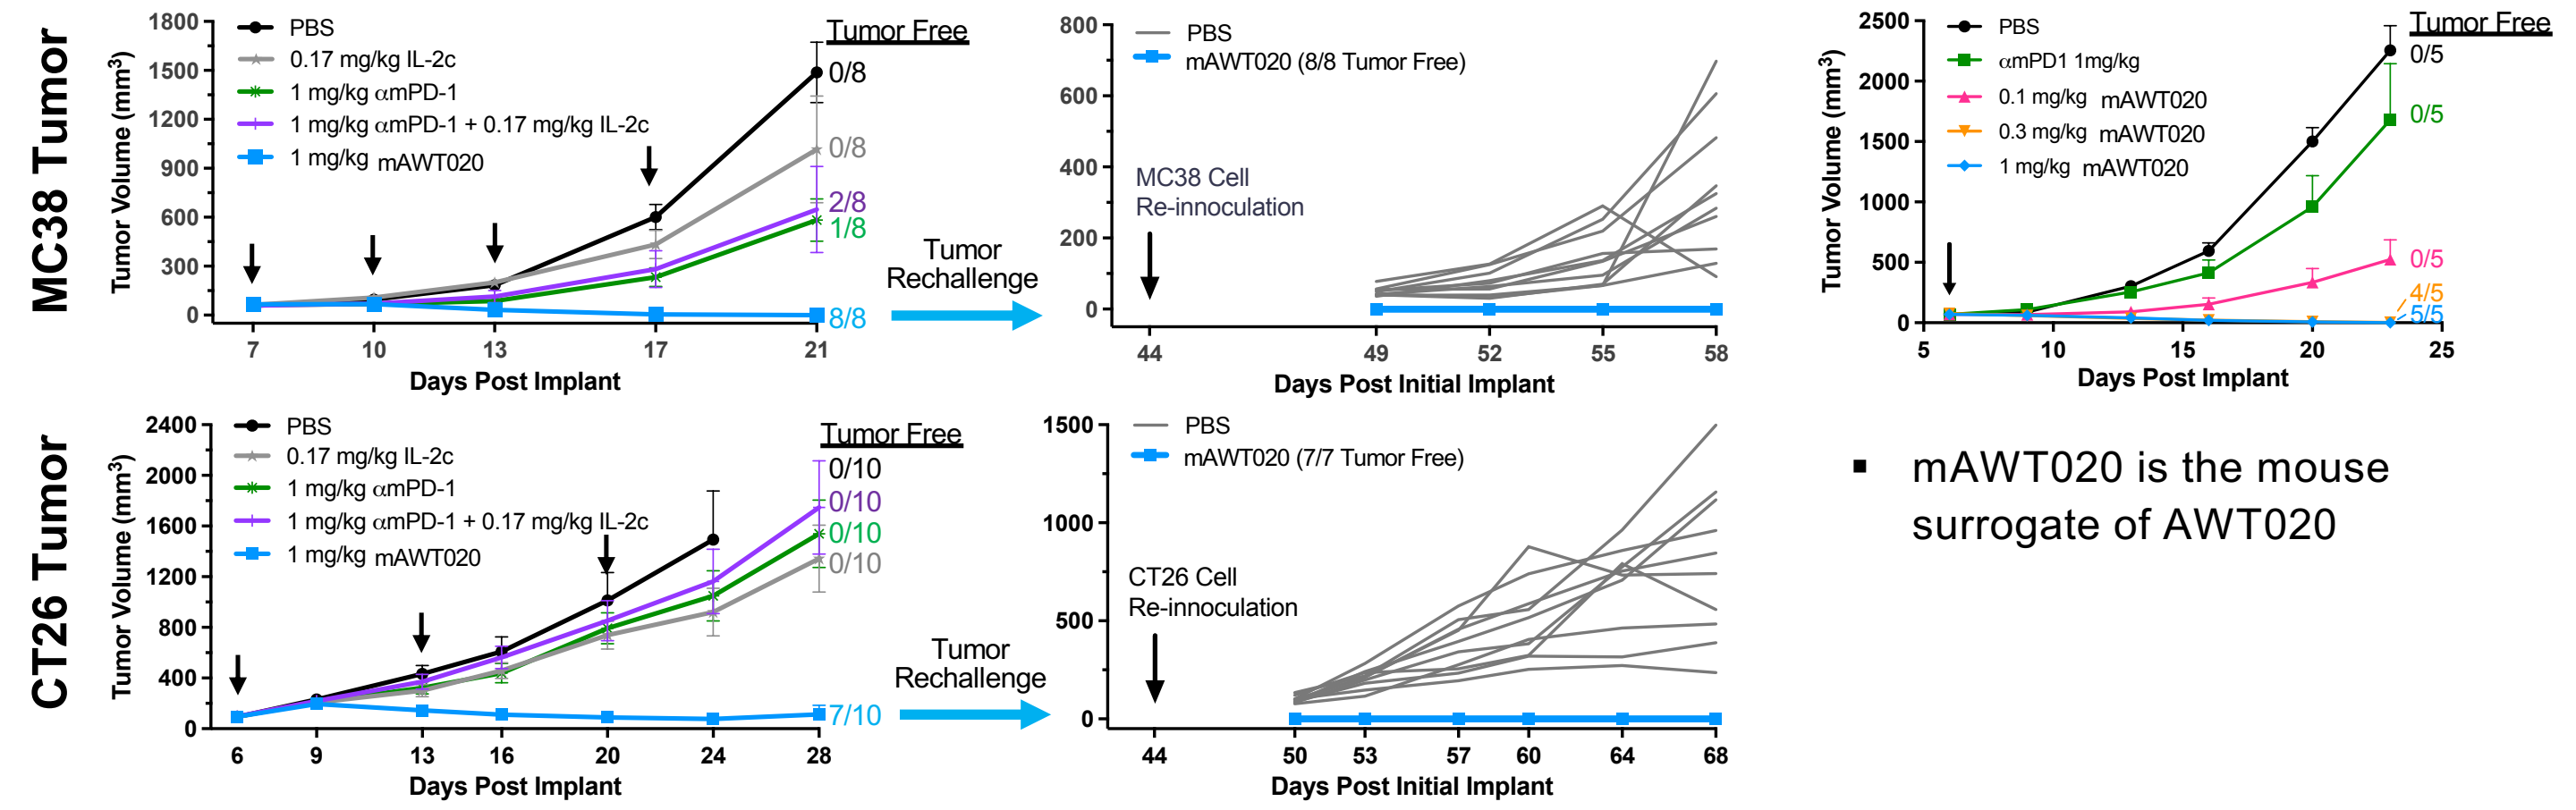
MC38 Tumor
CT26 Tumor
Tumor Volume (mm3)
PBS
0.17 mg/kg IL-2c
1 mg/kg αmPD-1
1 mg/kg αmPD-1 + 0.17 mg/kg IL-2c
1 mg/kg mAWT020
Tumor Free
Tumor Rechallenge
Days Post Implant
mAWT020 (8/8 Tumor Free)
MC38 Cell Re-innoculation
mAWT020 (7/7 Tumor Free)
CT26 Cell Re-innoculation
Days Post Initial Implant
αmPD1 1mg/kg
0.1 mg/kg mAWT020
0.3 mg/kg mAWT020
1 mg/kg mAWT020

- mAWT020 is the mouse surrogate of AWT020

mAWT020 Preferentially Expands and Activates T Cells in Tumor

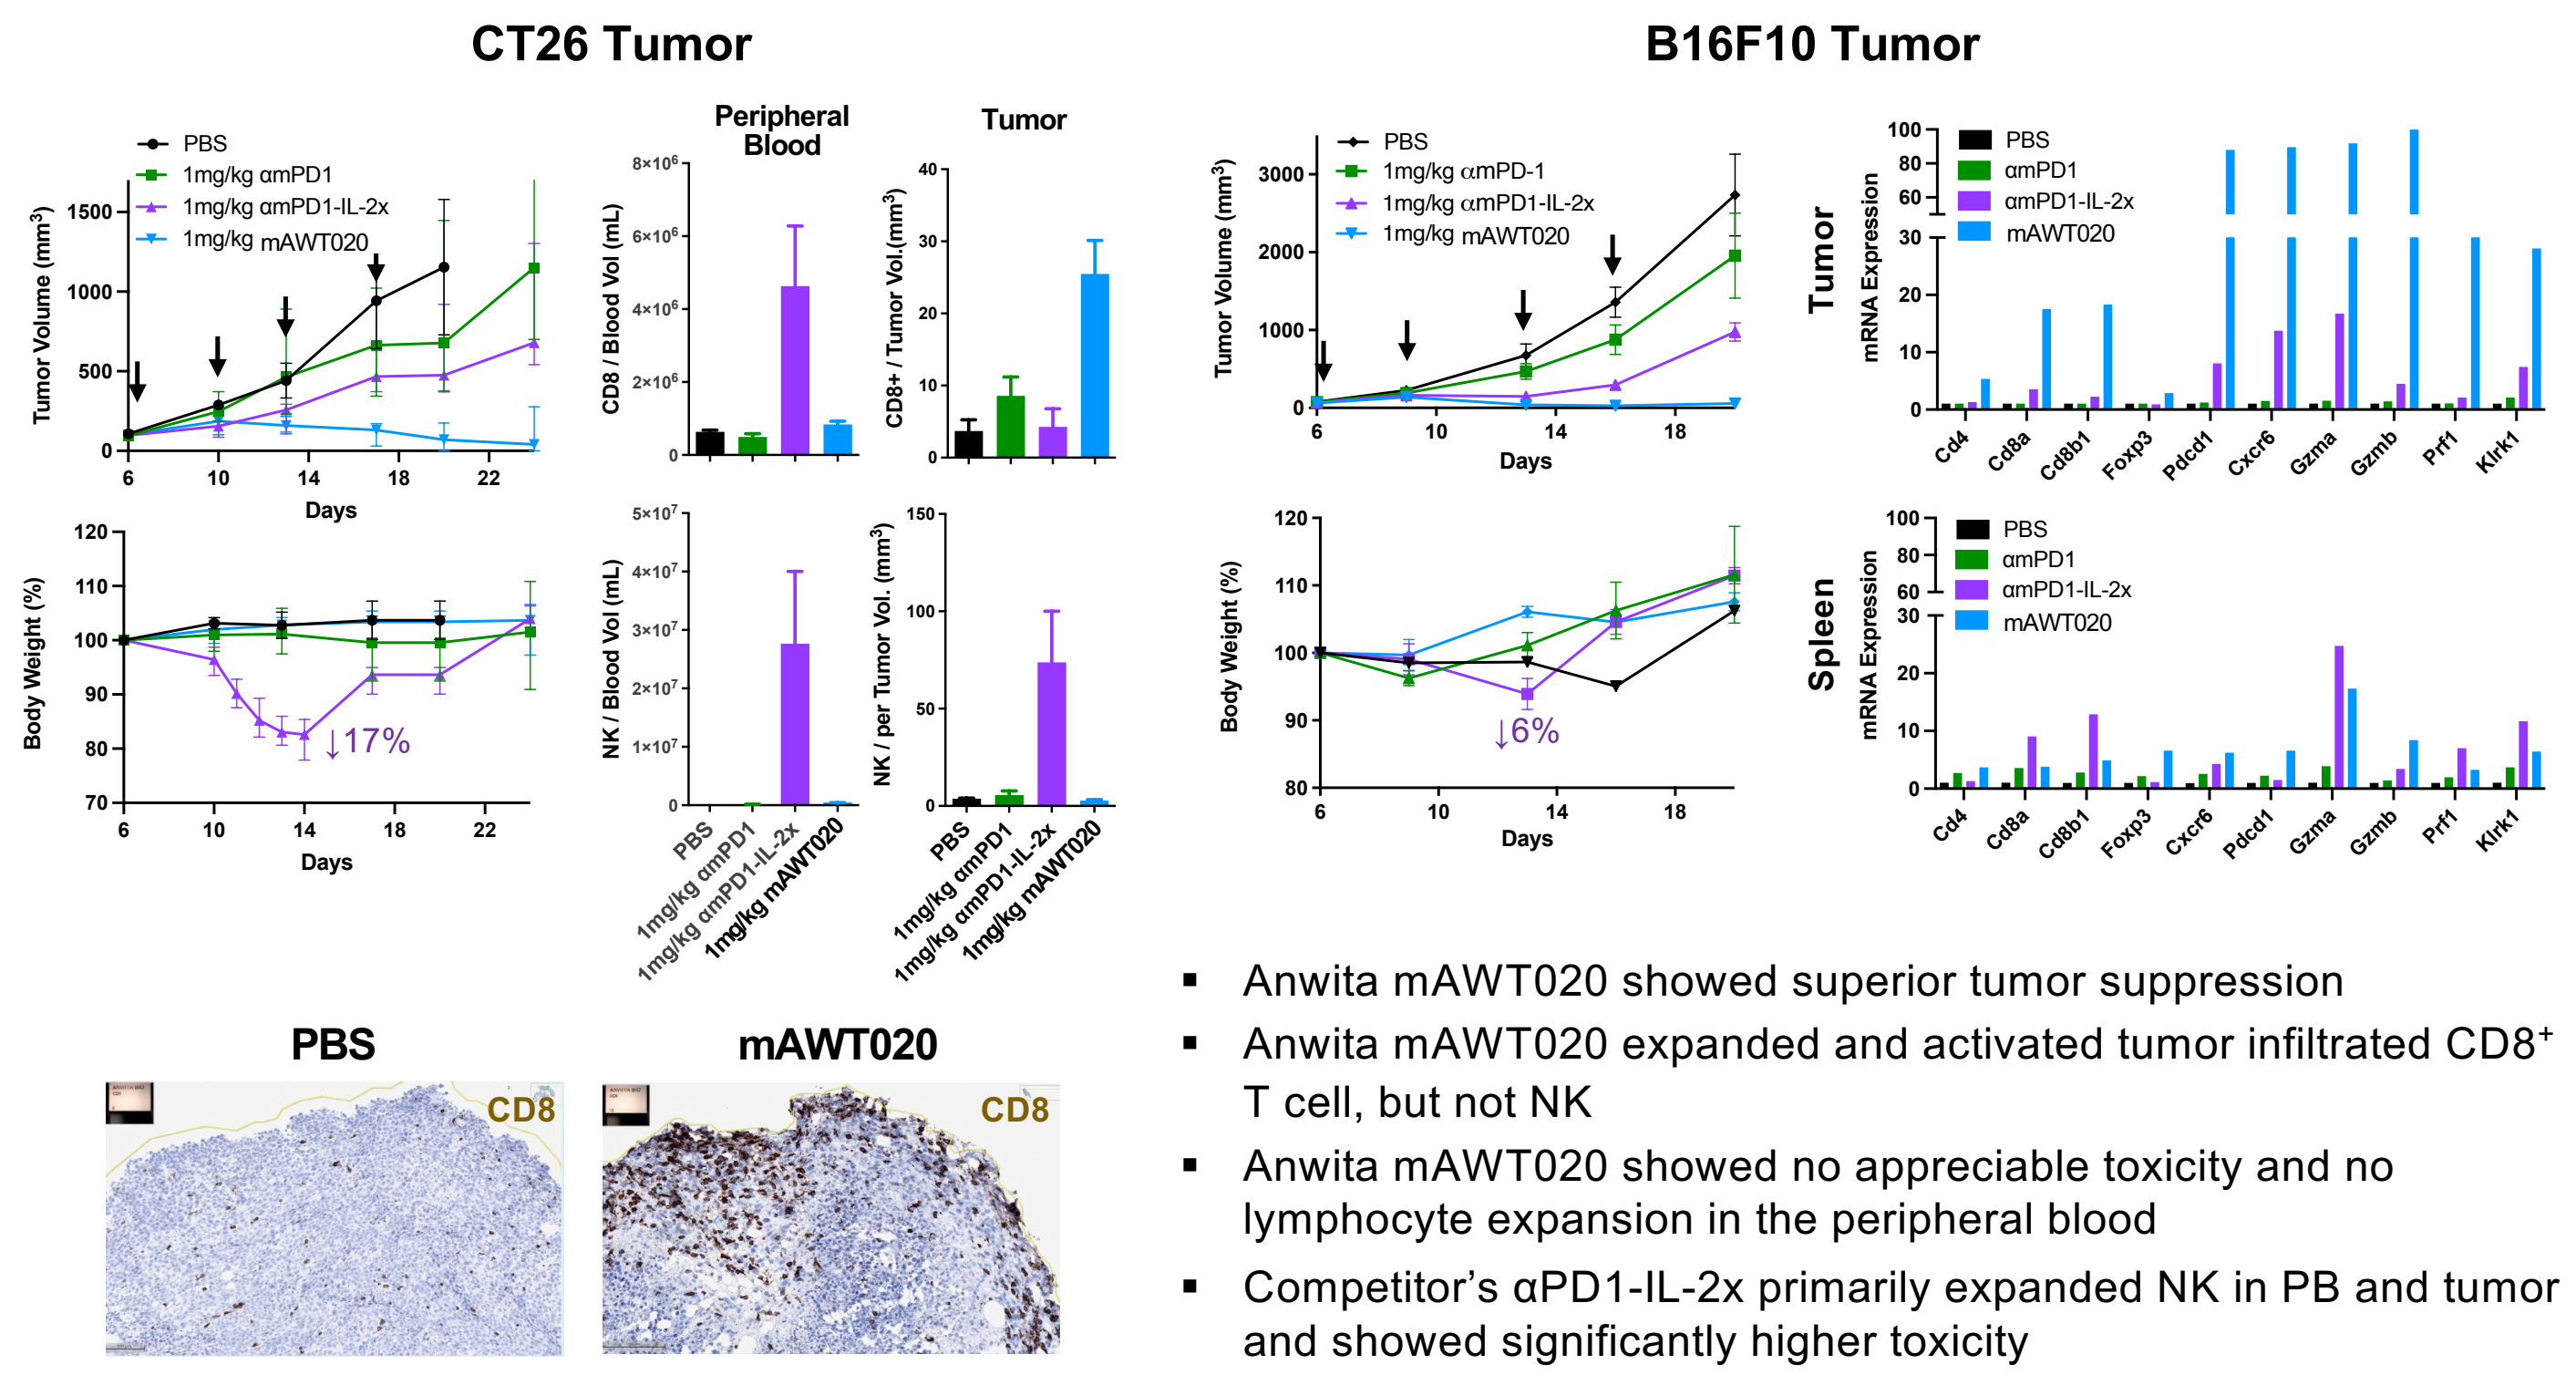
CT26 Tumor
B16F10 Tumor
PBS
1mg/kg αmPD1
1mg/kg αmPD1-IL-2x
1mg/kg mAWT020
Tumor Volume (mm3)
Body Weight (%)
Days
↓17%
↓6%
Peripheral Blood
Tumor
CD8 / Blood Vol (mL)
CD8+ / Tumor Vol.(mm3)
NK / Blood Vol (mL)
NK / per Tumor Vol. (mm3)
Spleen
mRNA Expression
Cd4
Cd8a
Cd8b1
Foxp3
Pdcd1
Cxcr6
Gzma
Gzmb
Prf1
Klrk1
PBS
mAWT020
CD8

- Anwita mAWT020 showed superior tumor suppression
- Anwita mAWT020 expanded and activated tumor infiltrated CD8+ T cell, but not NK
- Anwita mAWT020 showed no appreciable toxicity and no lymphocyte expansion in the peripheral blood
- Competitor's αPD1-IL-2x primarily expanded NK in PB and tumor and showed significantly higher toxicity

mAWT020 Avoids NK Driven Toxicity

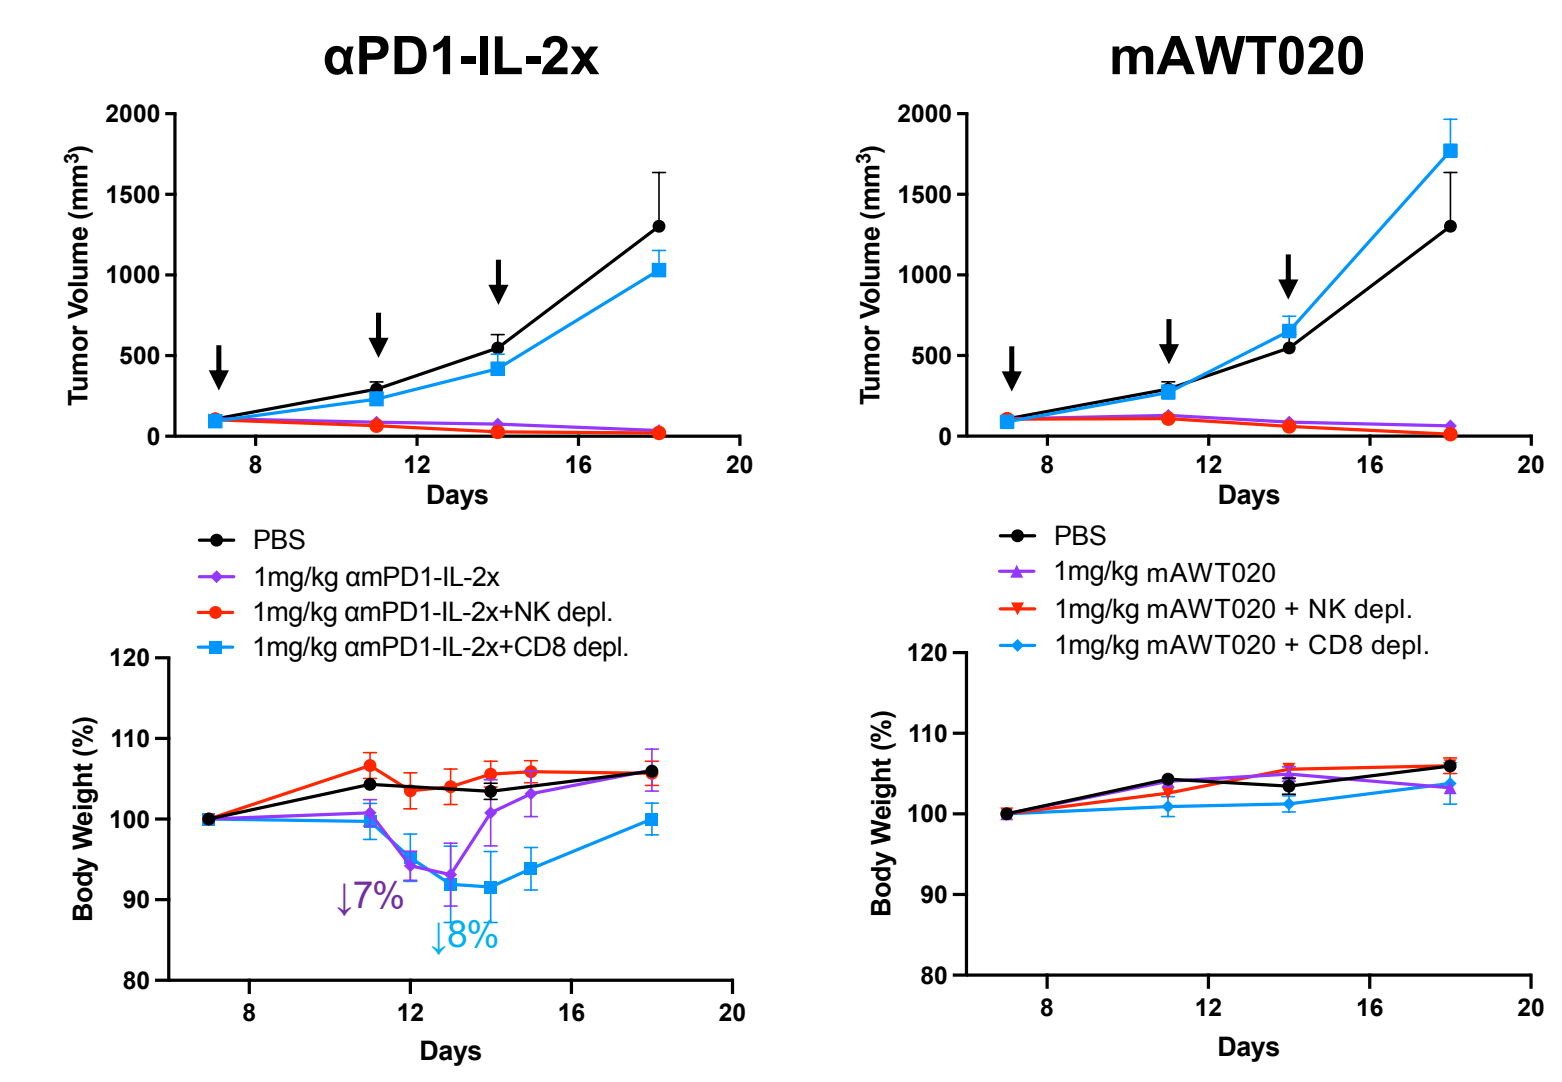
αPD1-IL-2x
mAWT020
Tumor Volume (mm3)
Body Weight (%)
Days
PBS
1mg/kg αmPD1-IL-2x
1mg/kg αmPD1-IL-2x+NK depl.
1mg/kg αmPD1-IL-2x+CD8 depl.
1mg/kg mAWT020
1mg/kg mAWT020 + NK depl.
1mg/kg mAWT020 + CD8 depl.
↓7%
↓8%

- To understand the roles of NK cell and CD8 T cell population, an in vivo depletion study was performed
- αNK antibody PK136 and αCD8 antibody Lyt 3.2 effectively depleted NK and CD8 T cell population respectively in C57/BL6 mice
- The antitumor efficacy of αPD1-IL-2 is mainly driven by CD8 T cell
- The toxicity of competitor's αPD1-IL-2x is mainly caused by NK cell
- Anwita's mAWT020 completely avoids NK driven toxicity

SUPERIOR SAFETY PROFILE

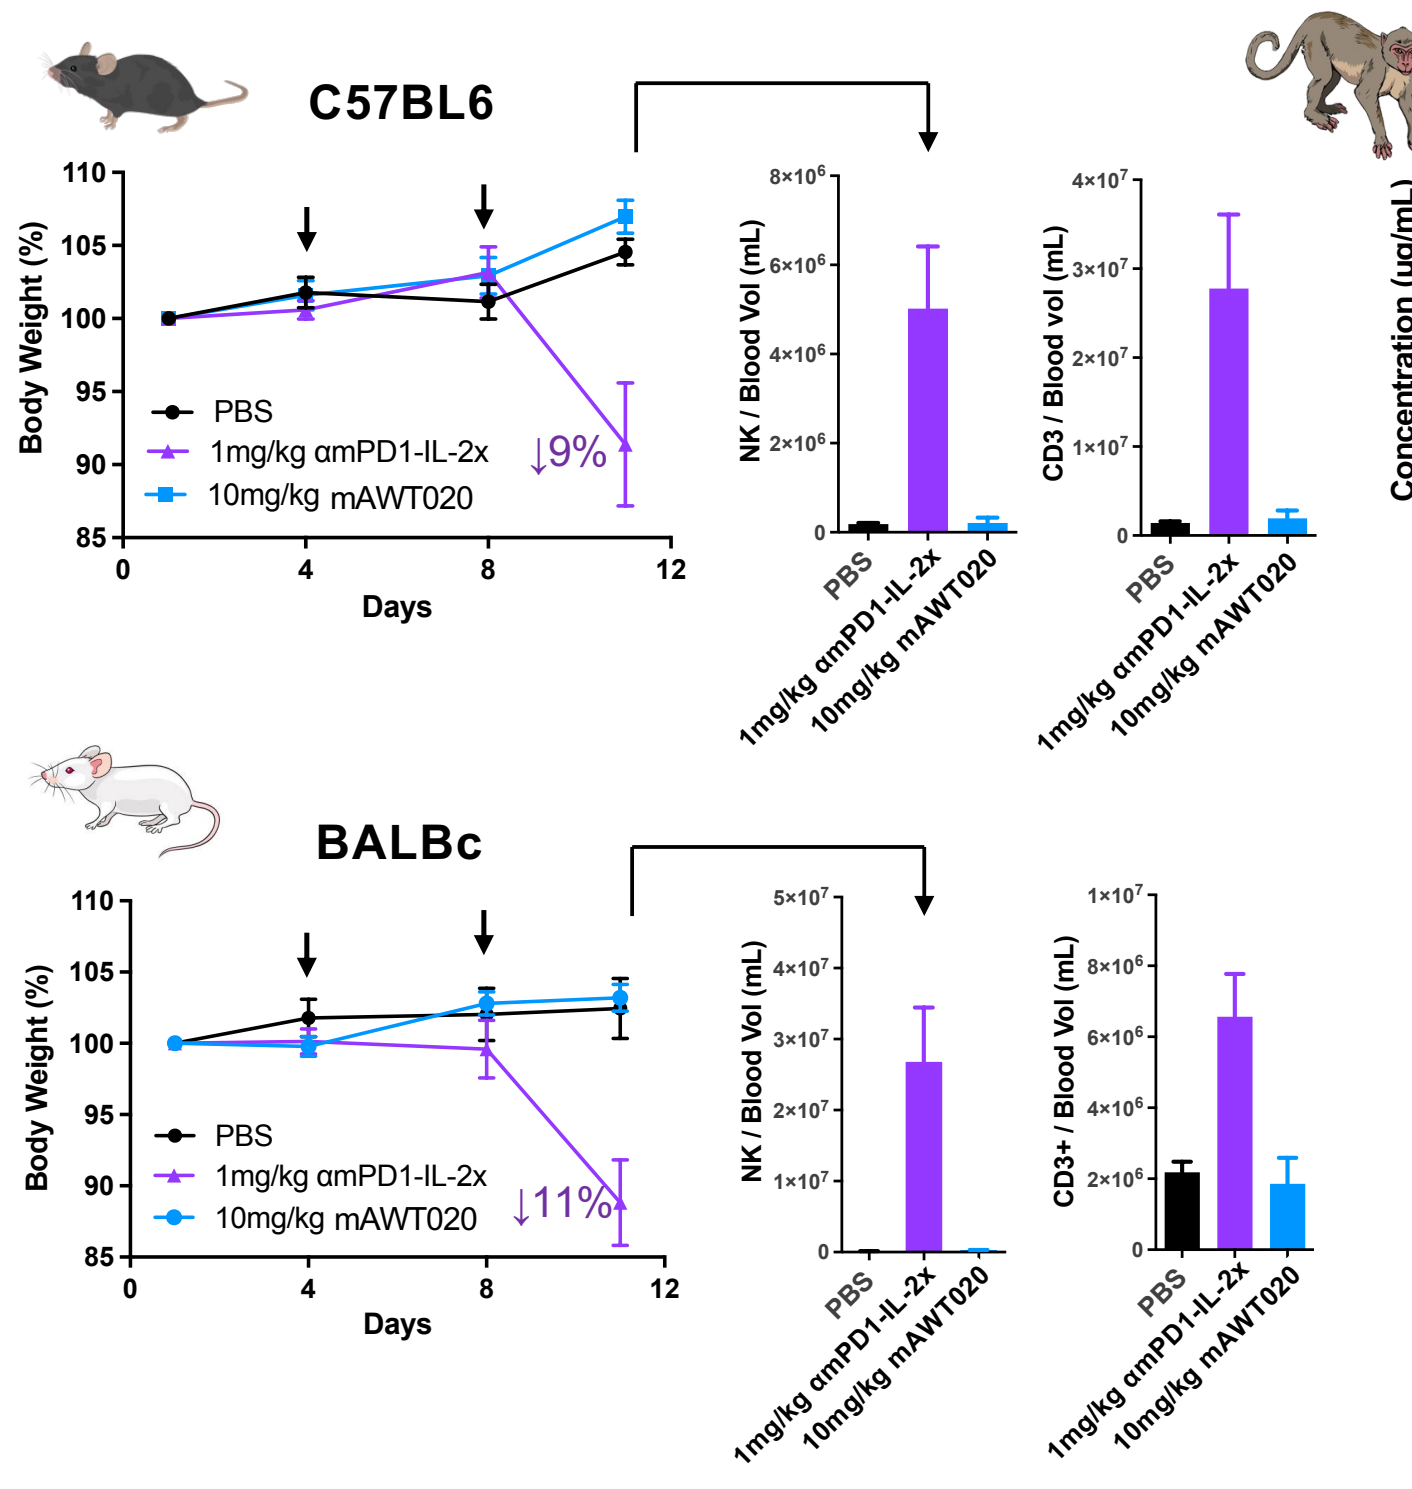
C57BL6
BALBc
Body Weight (%)
Days
PBS
1mg/kg αmPD1-IL-2x
10mg/kg mAWT020
↓9%
↓11%
NK / Blood Vol (mL)
CD3 / Blood vol (mL)
CD3+ / Blood Vol (mL)

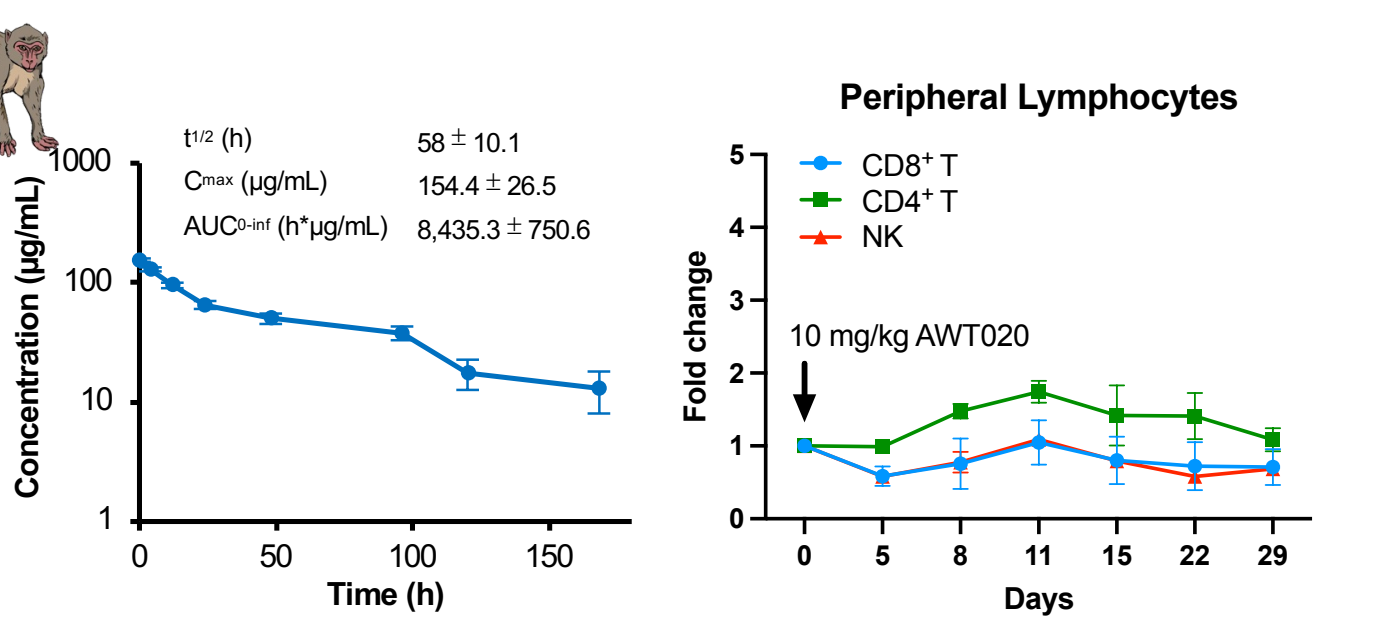
t1/2 (h) 58 ± 10.1
Cmax (μg/mL) 154.4 ± 26.5
AUC0-inf (h*μg/mL) 8,435.3 ± 750.6
Concentration (μg/mL)
Time (h)
Peripheral Lymphocytes
CD8+ T
CD4+ T
NK
Fold change
10 mg/kg AWT020
Days

In both C57BL6 and BALBc mice:

- 10 mg/kg of mAWT020 showed no NK expansion and no body weight drop

In cynomolgus monkey:

- AWT020 was well tolerated up to 10 mg/kg with prolonged half-life
- Minor change in peripheral lymphocytes, no significant increase of IL-6, IFN-g and TNF
- No cytokine release syndrome observed, no NK expansion

CONCLUSIONS

AWT020 is a Fully Optimized αPD1-IL2 for Superior Efficacy and Safety

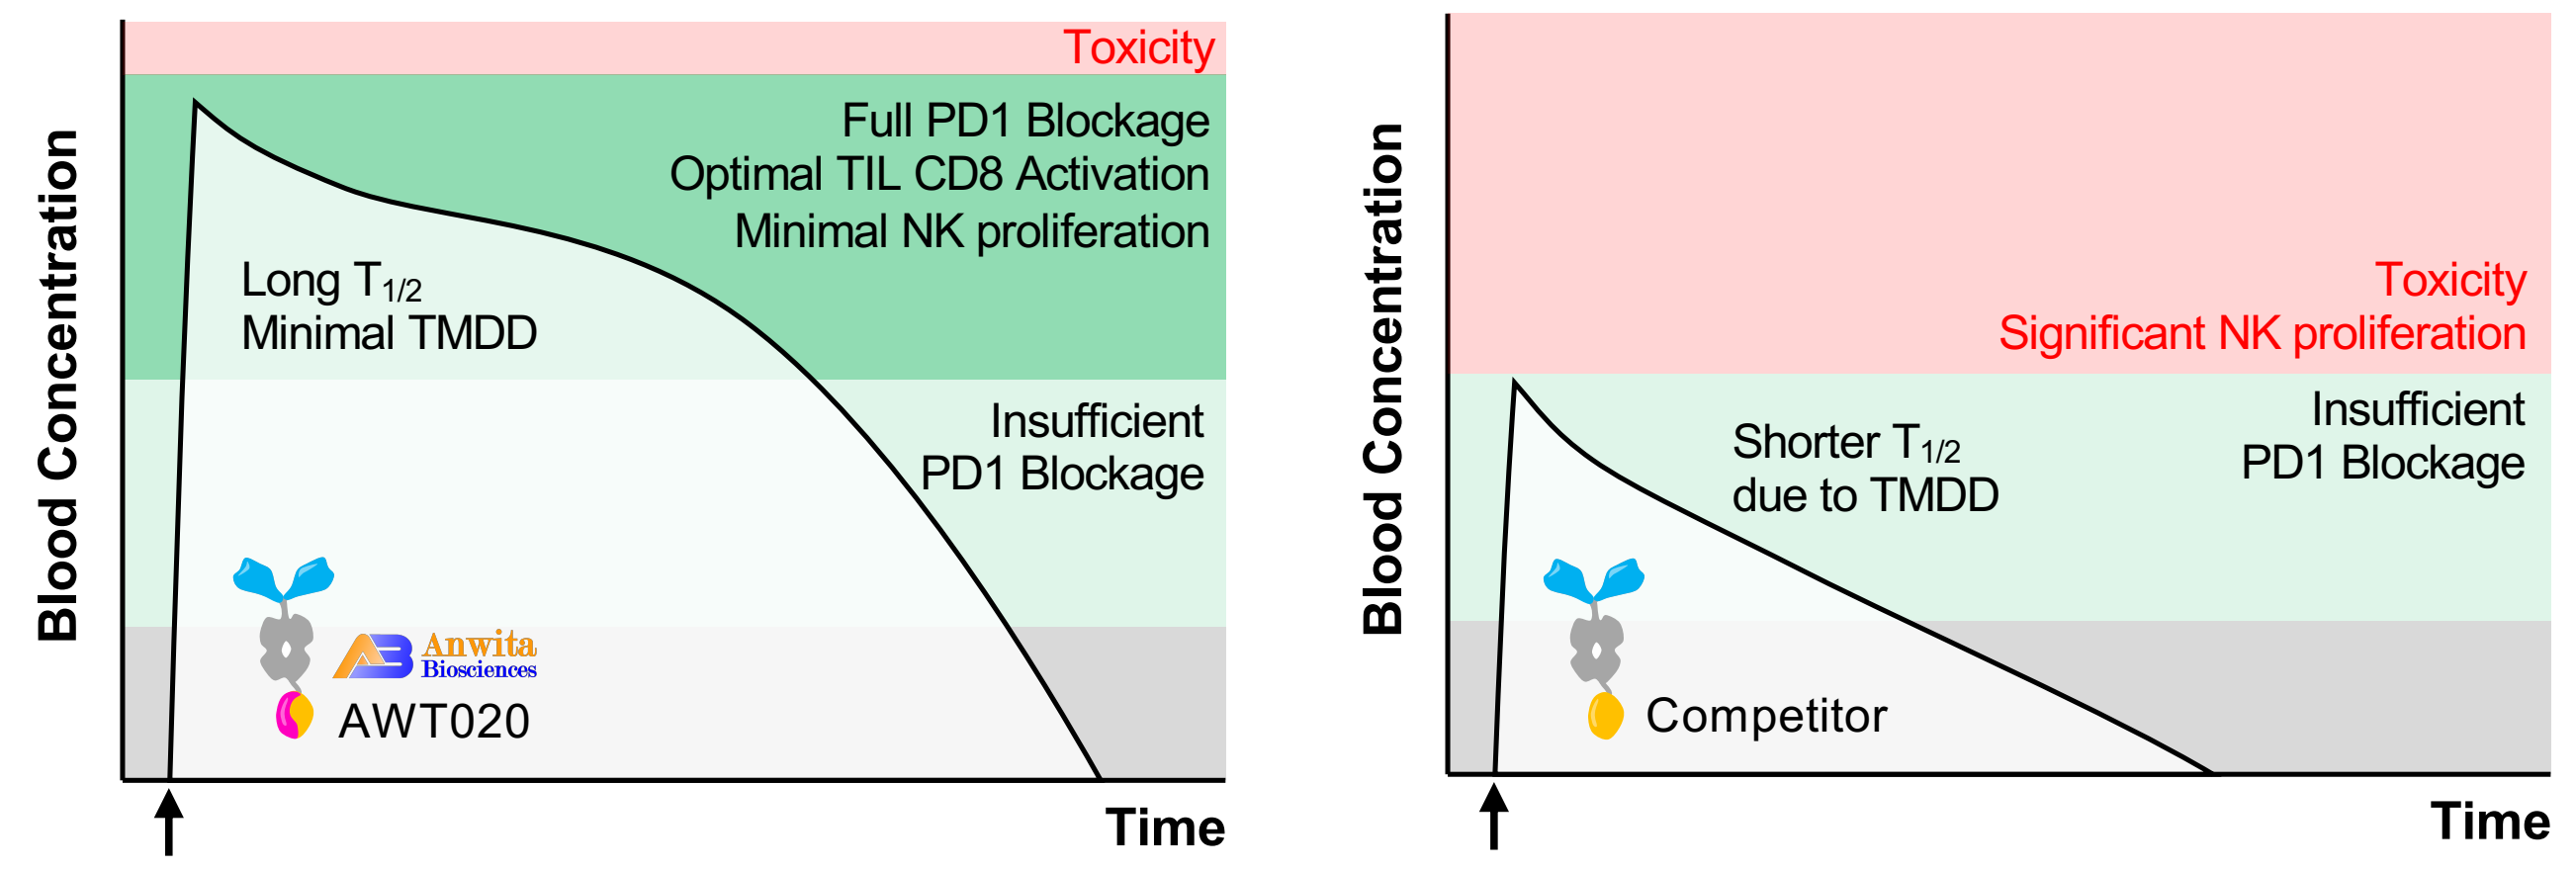
Toxicity
Full PD1 Blockage
Optimal TIL CD8 Activation
Minimal NK proliferation
Long T1/2
Minimal TMDD
Insufficient
PD1 Blockage
AWT020
Blood Concentration
Time
Toxicity
Significant NK proliferation
Shorter T1/2
due to TMDD
Insufficient
PD1 Blockage
Competitor

		AWT020 Anwita	αPD1-IL2x Leading competitor
Activity and Efficacy	CD8 T expansion in PB	<2x	~4x
	CD8 T expansion in Tumor	>5x	<2x
	NK expansion in PB	<2x	>20x
	NK expansion in Tumor	<2x	>10x
	Anti-Tumor Efficacy (TGI)	Superior	Good
Toxicity and PK	MTD (Mouse)	>10 mg/kg	~1 mg/kg
	MTD (Monkey)	>10 mg/kg	est. ~1 mg/kg
	PK (Monkey)	Superior, minimal TMDD	Strong TMDD
	Therapeutic Window	>100	est. ~3-10

In summary, the preclinical observations support the development of AWT020 as revolutionary IL-2 immunotherapy for cancer. An Investigational New Drug (IND) application is planned for AWT020 by 1H 2023.

References
1. Jiang et al. 2016
2. Clark et al. 2021
3. Rosenberg et al. 2014
4. Dutcher et al. 2014
5. Krieg et al. 2010
6. Hu et al. 2003
7. Ptacin et al. 2021

Download this poster →